Novel Humanized Particles for Efficient Delivery of CRISPR and Other Gene Editors

Peter K. Cabeceiras[1,2,3,4,5], Noah Adams[1], Tian Duo Wang[2,3,5,6,*], Christopher Woodilla[2,3,5], Caitlin Crandall[1], Bevan Bsharah[1], Xiaozhi Ren[1], Yanjing Li[1], Andrew Kane[1], Jeffrey Lee[1], Molly Riley[1], Annie Elliott[1], Vy Phan[1], Carrie Ng[7,8], Cosmin Mihai[1], Jason Auer[1], Casey A. Maguire[7,8], Melissa Bonner[1], and J. Keith Joung[2,3,4,5,6,*]

[1]Nvelop Therapeutics, Cambridge, MA, [2]Molecular Pathology Unit, Massachusetts General Hospital, Charlestown, MA, [3]Center for Cancer Research, Massachusetts General Hospital, Charlestown, MA, [4]Program in Biological and Biomedical Sciences, Harvard Medical School, Boston, MA, [5]Center for Computational and Integrative Biology, Massachusetts General Hospital, Charlestown, MA, [6]Department of Pathology, Harvard Medical School, Boston, MA, [7]Department of Neurology, Massachusetts General Hospital, Boston, MA, [8]Harvard Medical School, Boston, MA,
[*]Current address: Arena BioWorks LLC, Cambridge, MA

Introduction

DLVR-M, pictured here, is a novel platform for simplified protein delivery based on an envelope-pseudotyped extracellular vesicle encapsulating a cargo fused to a pleckstrin homology (PH) domain.[1]

- DLVR-M particles pseudotyped with different novel envelope proteins possess varied cell-type tropisms.
- DLVR-M particles can deliver CRISPR gene editing nucleases, base editors and prime editors into a broad range of therapeutically relevant primary human cells (including HSPCs, neurons, and skeletal muscle) and induce highly efficient editing of >80%.

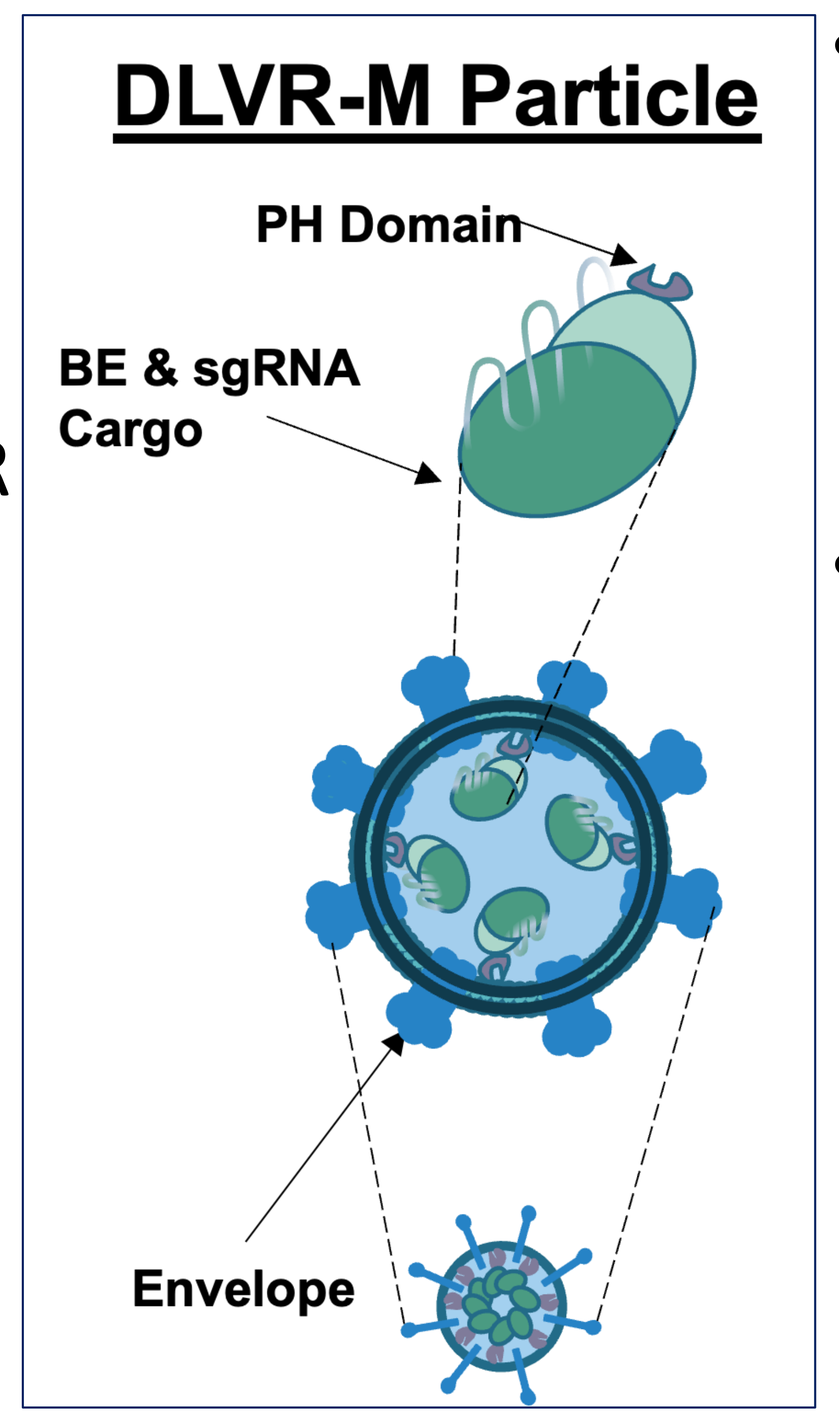

DLVR-M Particle
PH Domain
BE & sgRNA Cargo
Envelope

- DLVR-M particles can induce robust and efficient *in vivo* adenine base editing in mouse liver on a therapeutic gene target (*PCSK9*).
- Humanized DLVR-M particles, made without using any viral proteins, can efficiently package and deliver gene editors, raising the potential for reduced immunogenicity and redosing with this platform.

Results

RNP Delivery Platform — Figure 1. DLVR-M particles match or outperform virus-like particles (VLPs) for delivery of Cas9 nucleases to human cells

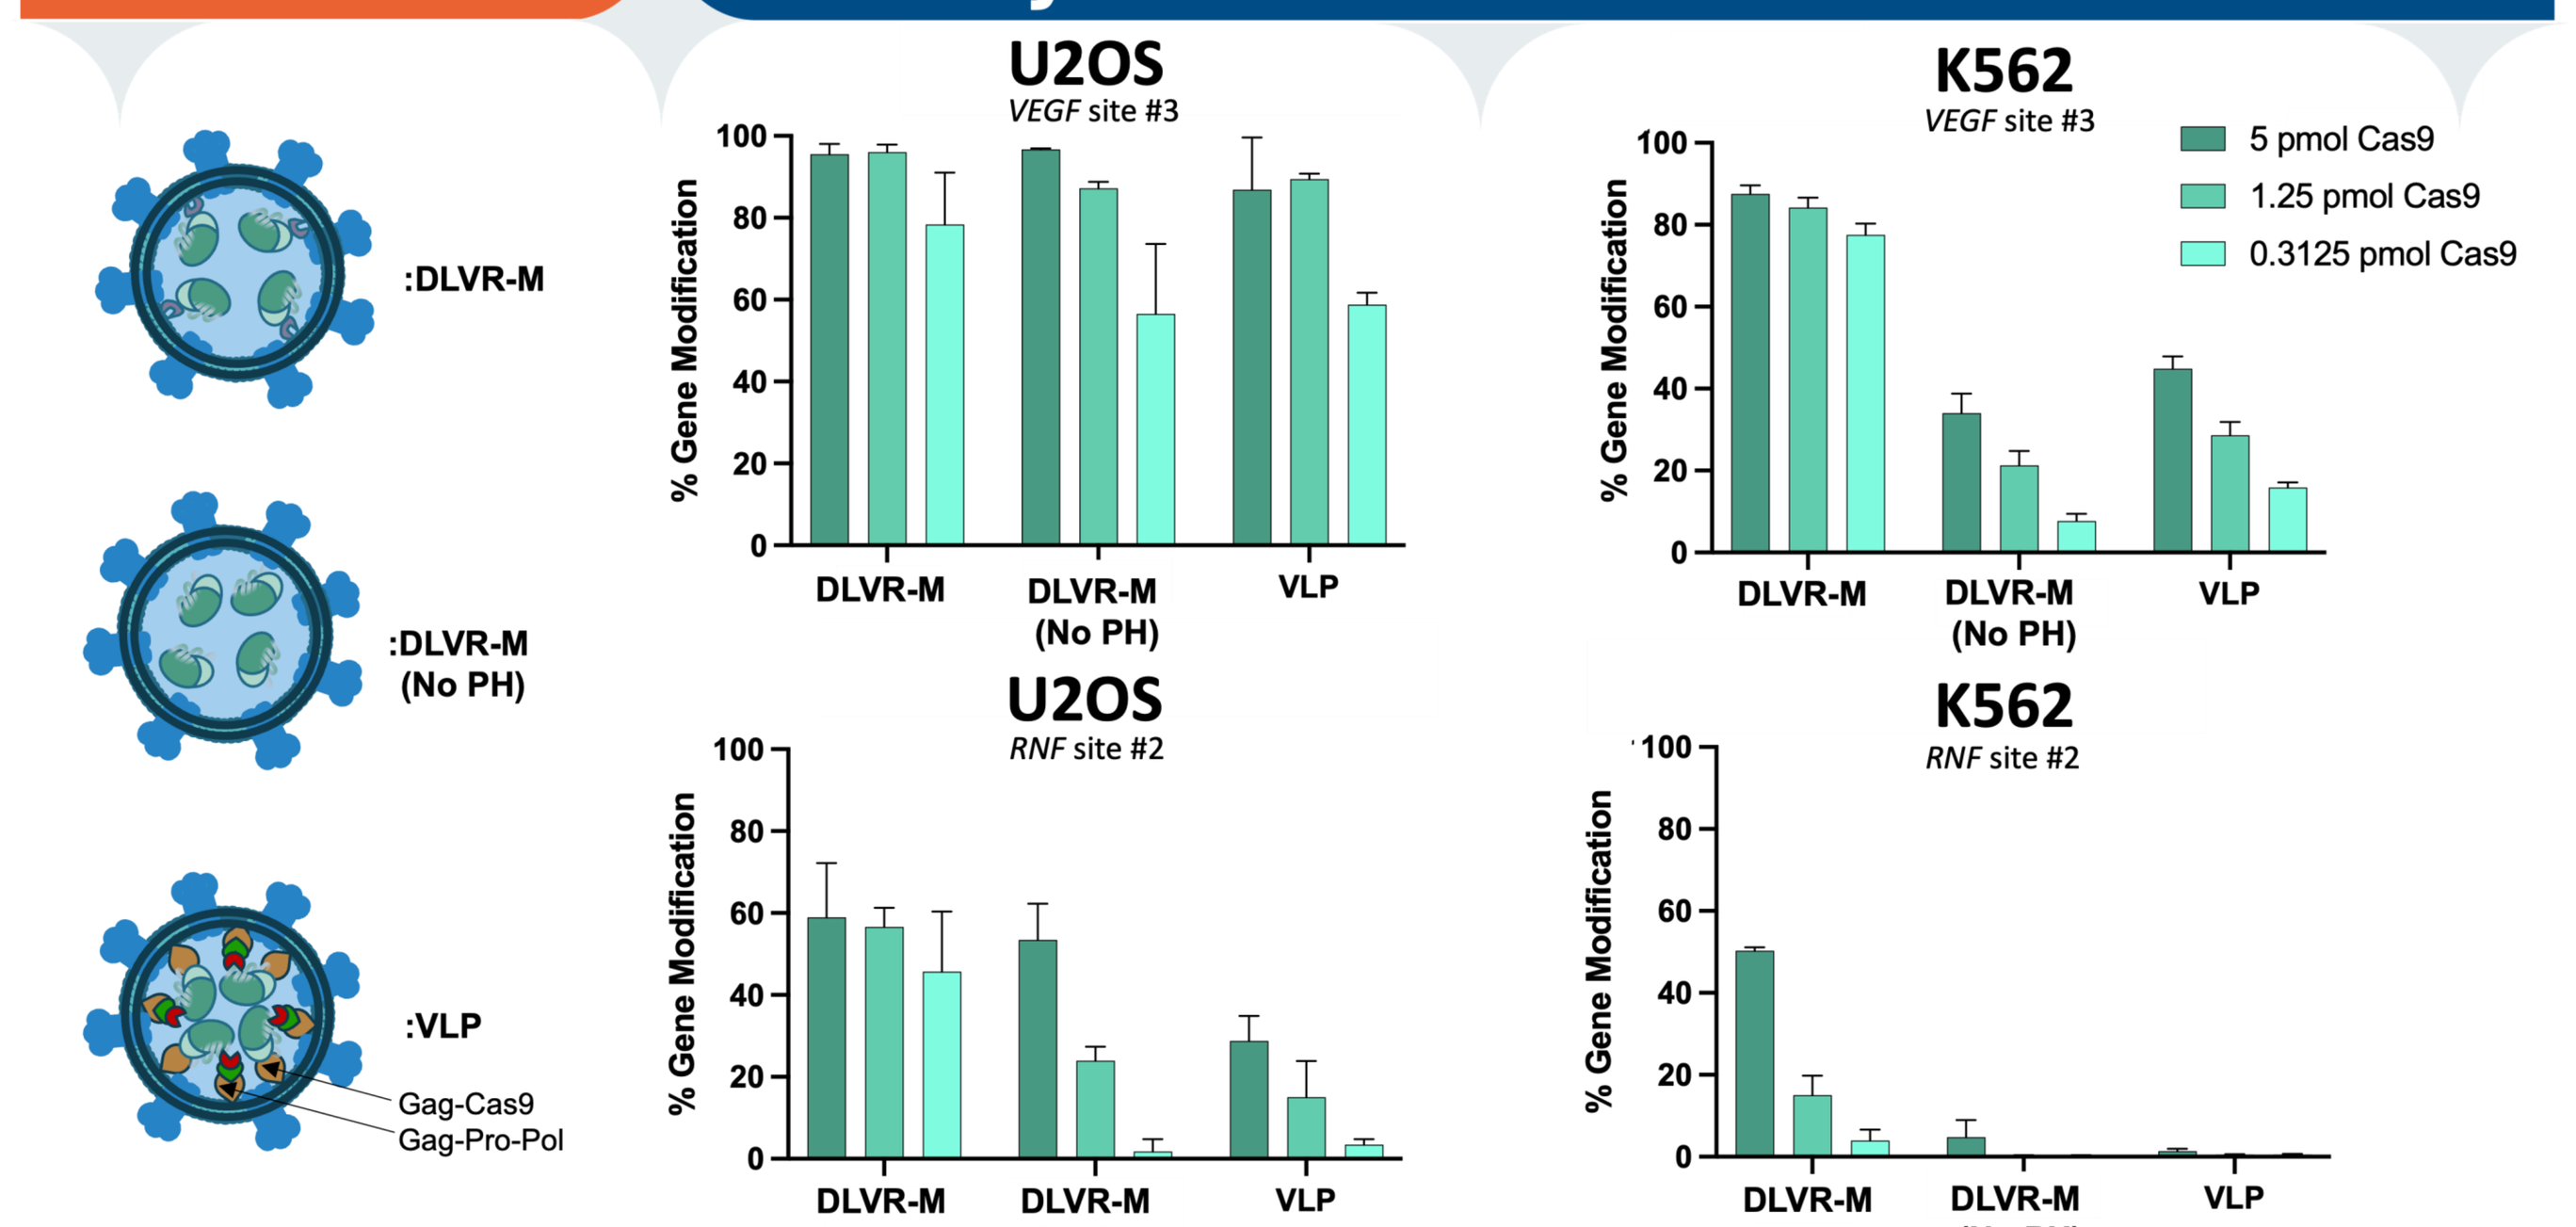

:DLVR-M
:DLVR-M (No PH)
:VLP
Gag-Cas9
Gag-Pro-Pol
U2OS
VEGF site #3
K562
VEGF site #3
5 pmol Cas9
1.25 pmol Cas9
0.3125 pmol Cas9
U2OS
RNF site #2
K562
RNF site #2
% Gene Modification
DLVR-M
DLVR-M (No PH)
VLP

Particles were concentrated 100x by polyethylene glycol (PEG) precipitation. Cas9 concentration was determined by enzyme-linked immunosorbent assay (ELISA). All delivery platforms, including VLPs,[2] were pseudotyped with VSVG and normalized on the basis of Cas9 prior to transduction of two cell types (U2OS-more delivery permissive & K562-less delivery permissive). *VEGF* site #3 and *RNF* site #2 are relatively high and low activity editing sites, respectively. Gene editing was quantified by next generation sequencing (NGS).

DLVR-M — Figure 2. DLVR-M PH domain enhances delivery across different types of cells

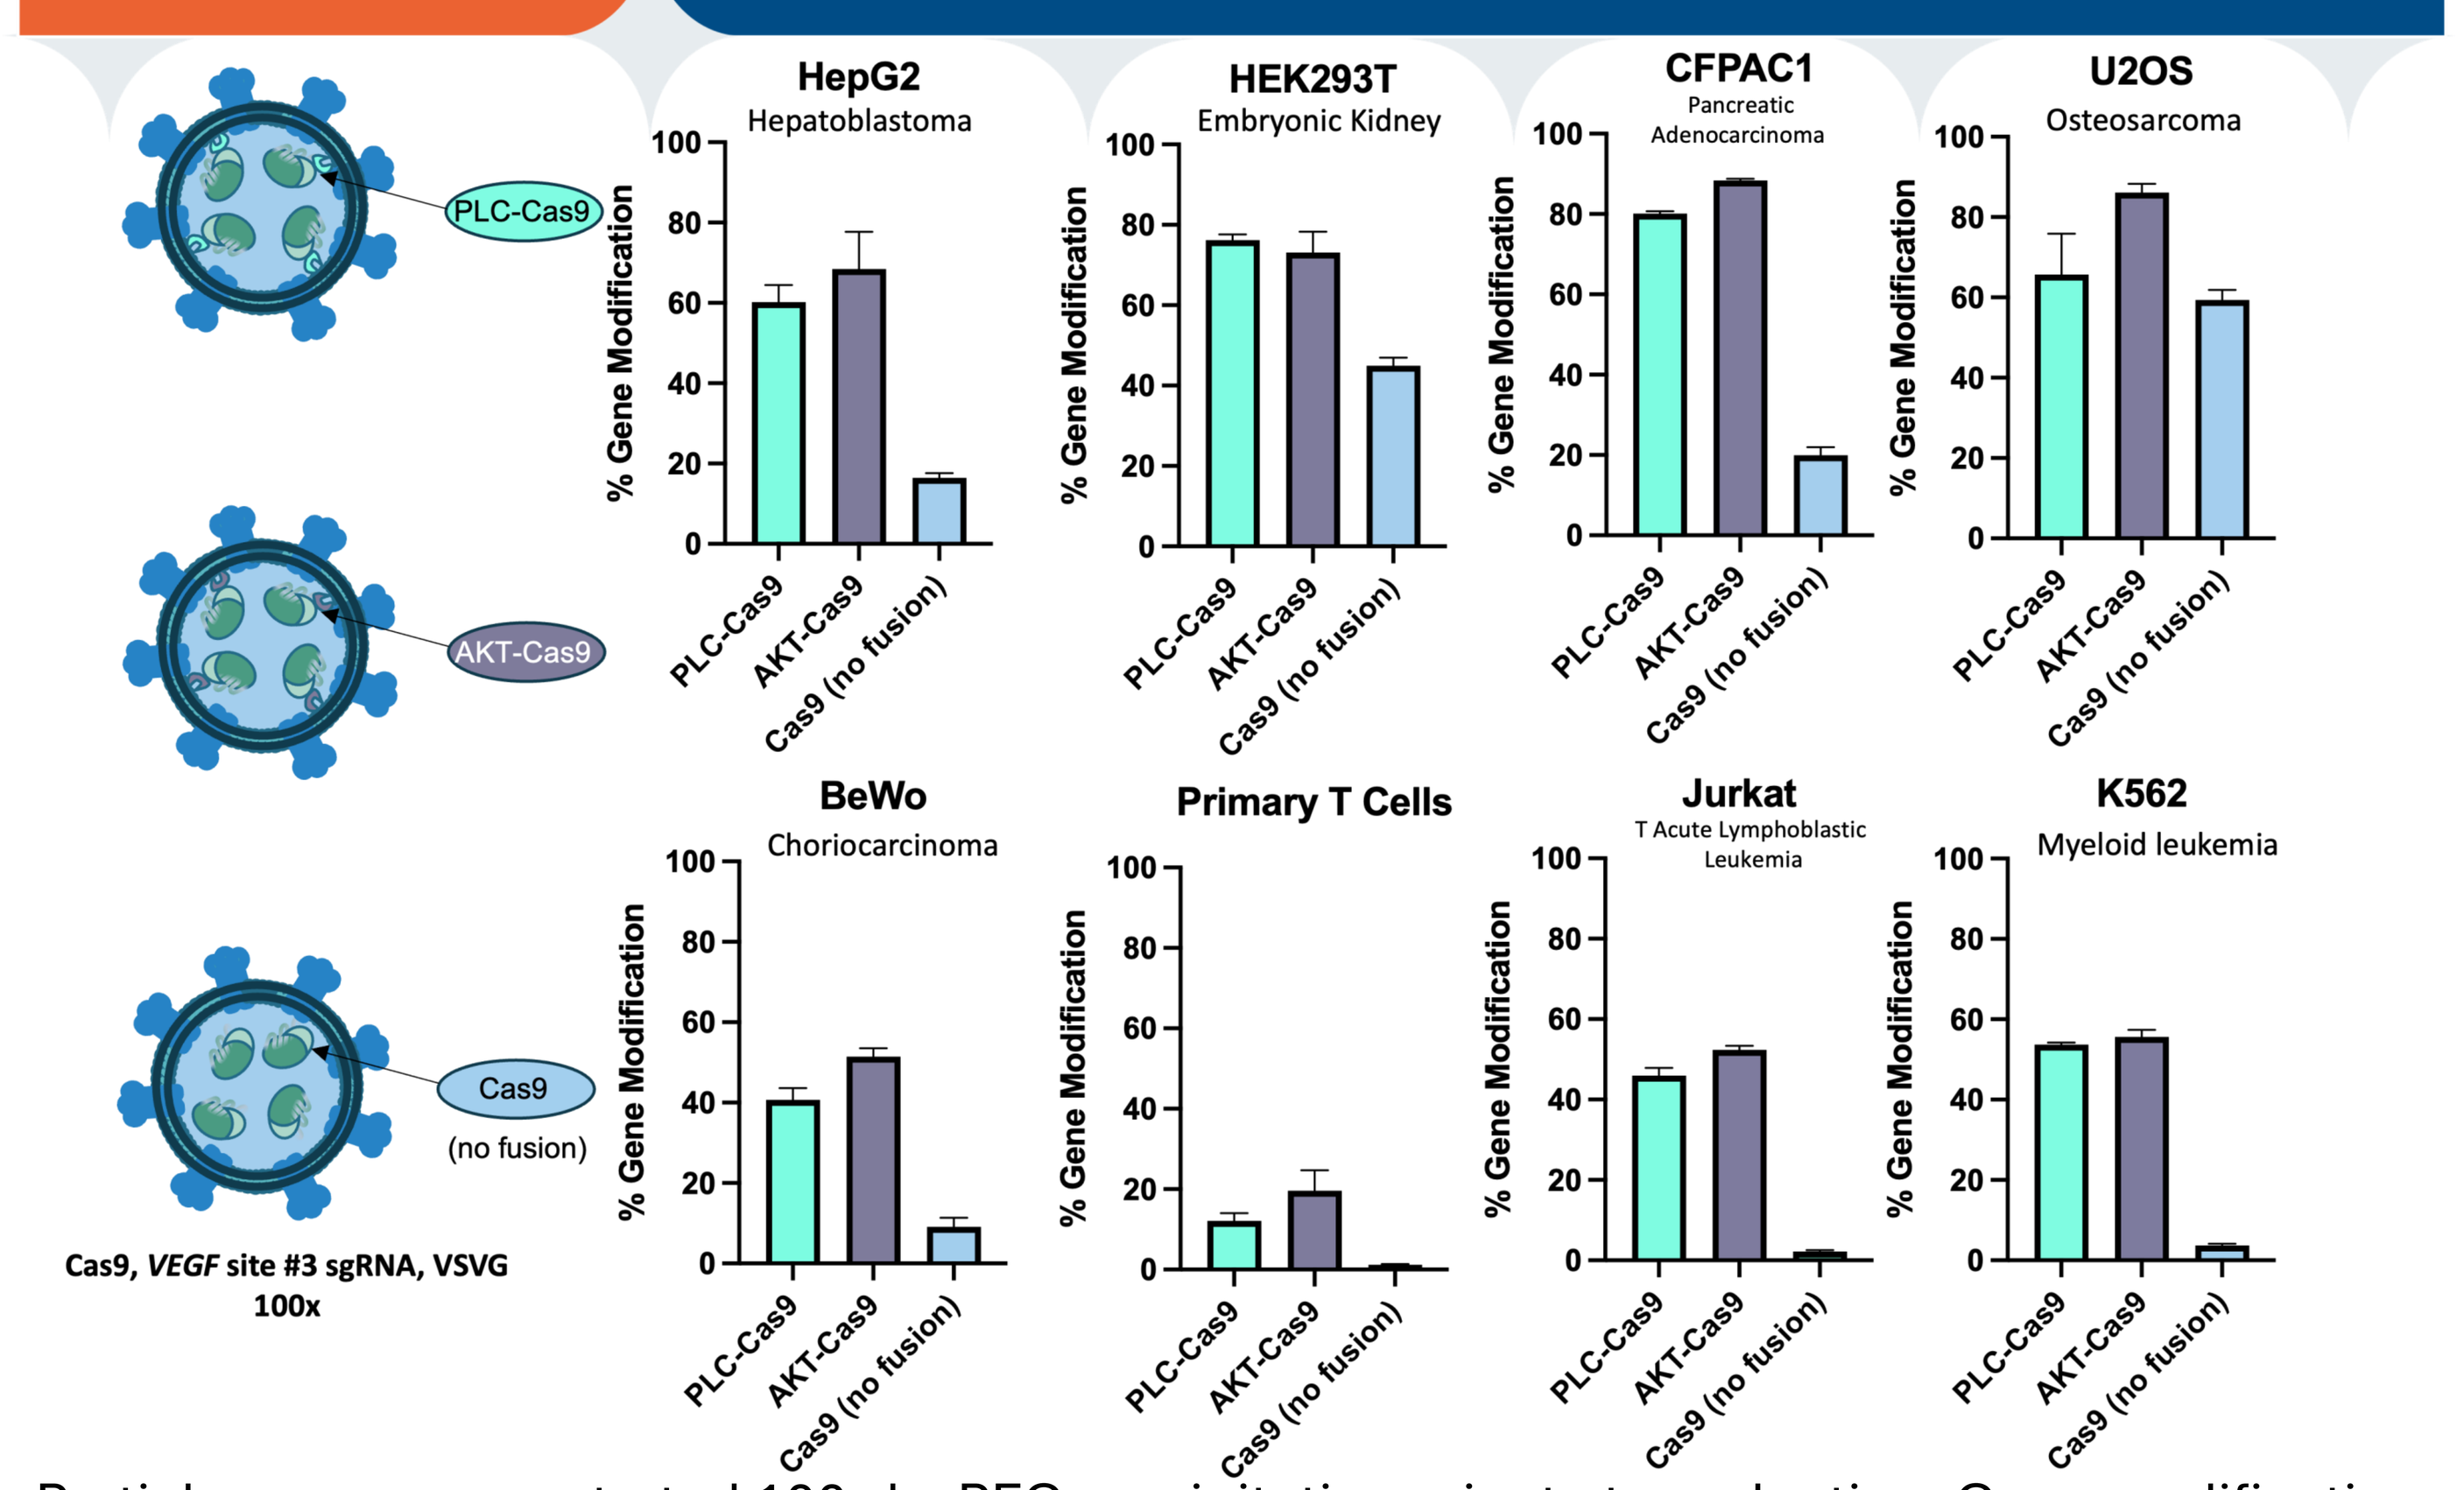

PLC-Cas9
AKT-Cas9
Cas9 (no fusion)
Cas9, VEGF site #3 sgRNA, VSVG 100x
HepG2 Hepatoblastoma
HEK293T Embryonic Kidney
CFPAC1 Pancreatic Adenocarcinoma
U2OS Osteosarcoma
BeWo Choriocarcinoma
Primary T Cells
Jurkat T Acute Lymphoblastic Leukemia
K562 Myeloid leukemia
% Gene Modification

Particles were concentrated 100x by PEG precipitation prior to transduction. Gene modification % was quantified by NGS.

DLVR-M PH Screen — Figure 3. DLVR-M PH screen yields optimized architecture

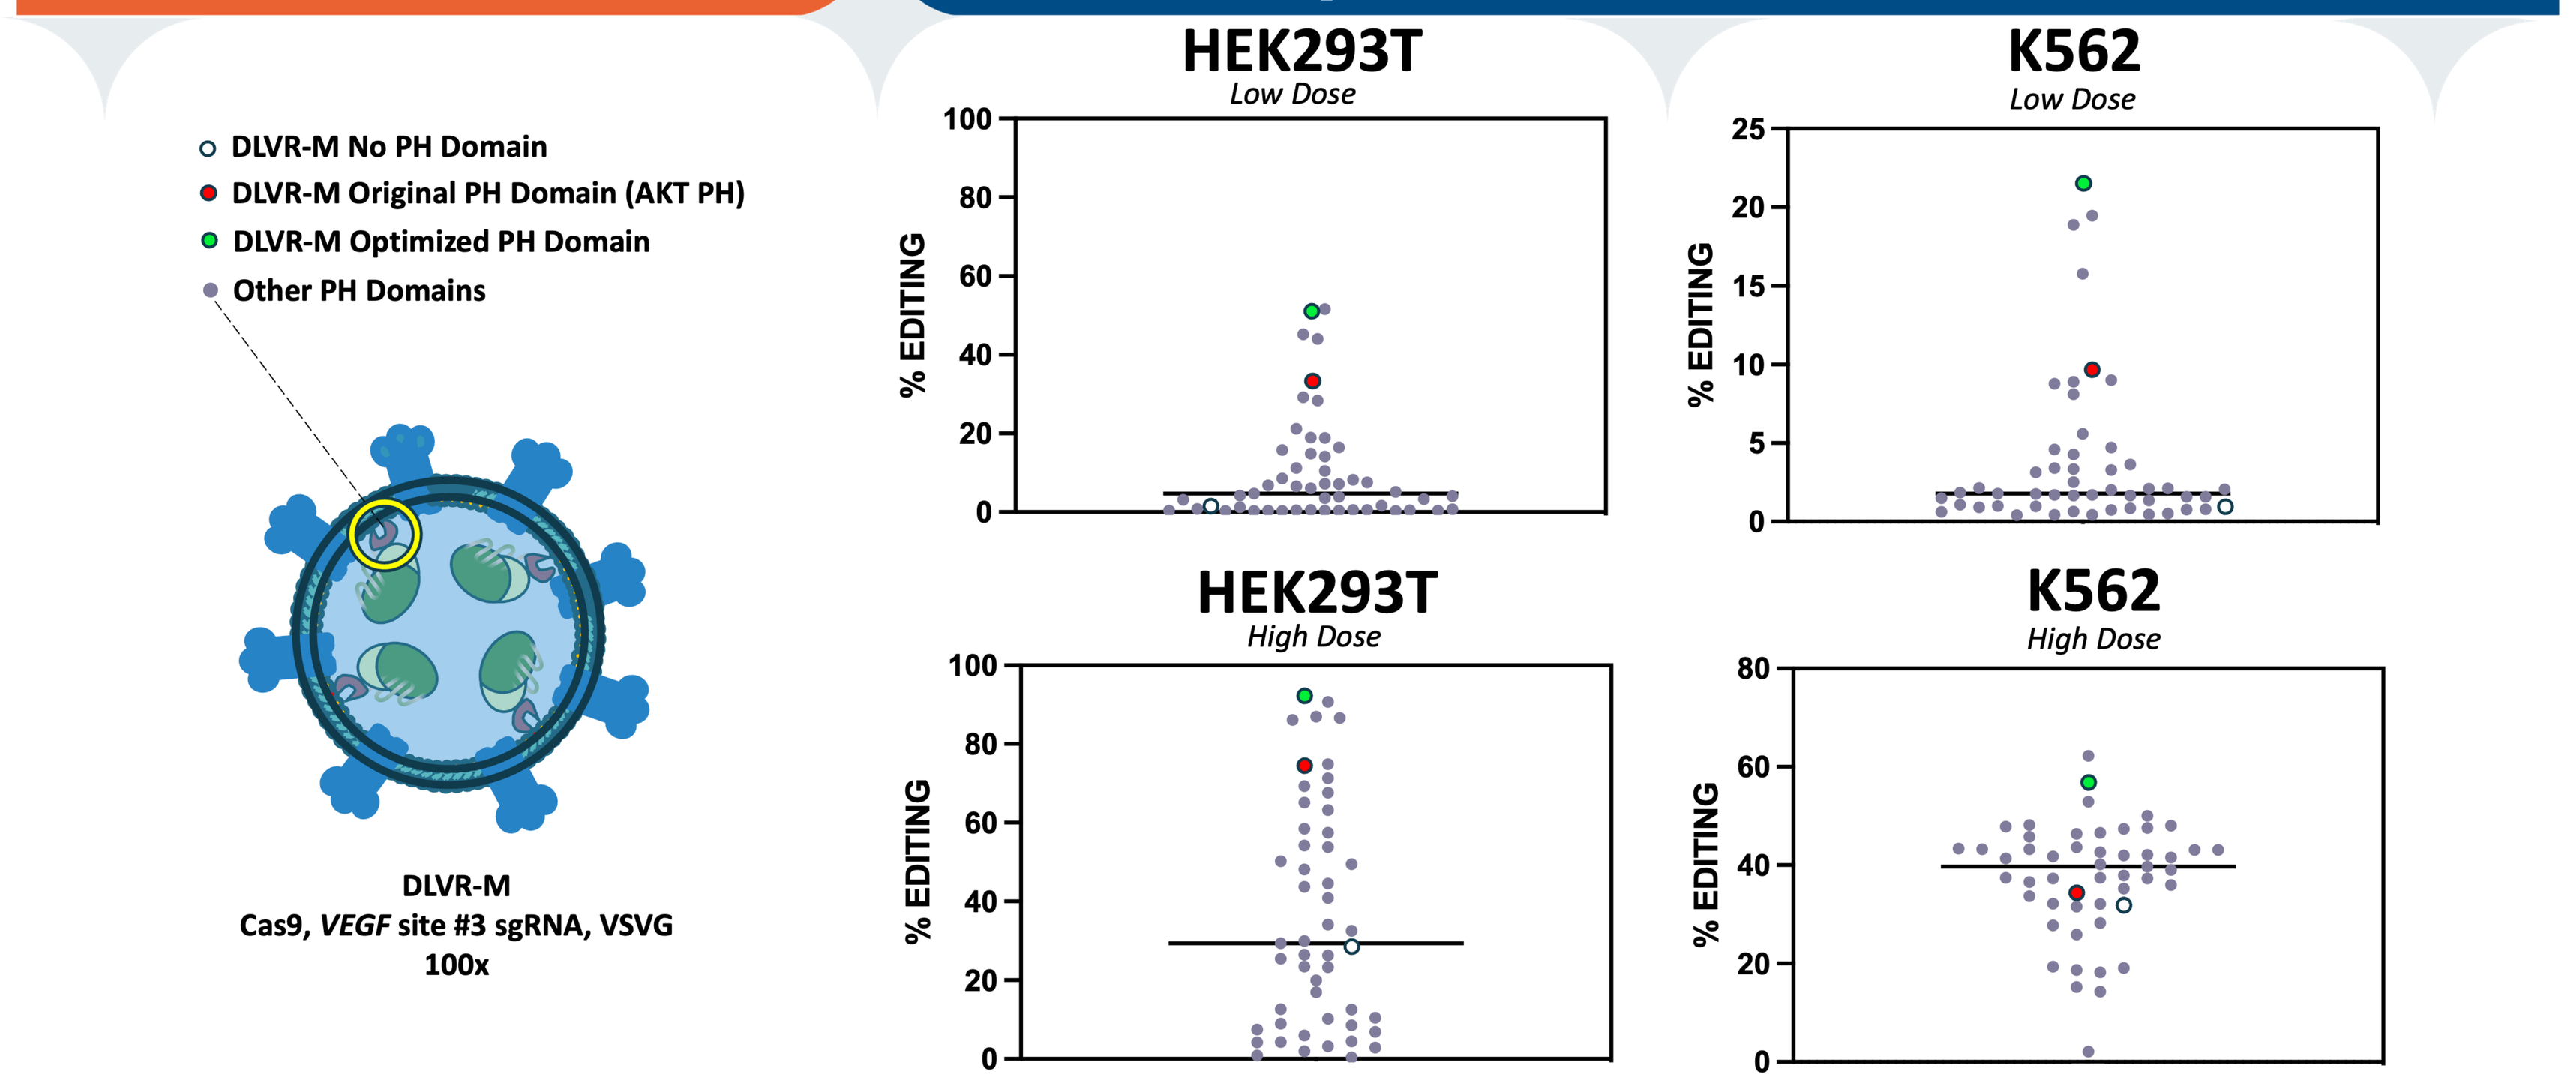

DLVR-M No PH Domain
DLVR-M Original PH Domain (AKT PH)
DLVR-M Optimized PH Domain
Other PH Domains
DLVR-M
Cas9, VEGF site #3 sgRNA, VSVG
100x
HEK293T Low Dose
K562 Low Dose
HEK293T High Dose
K562 High Dose
% EDITING

50 PH domain fusions to Cas9 were tested across two different recipient cell types at low and high doses. Particles were concentrated 100x by PEG precipitation prior to transduction of recipient cells. The top performing PH domain across recipient cells and doses was incorporated into the DLVR-M architecture and used in all subsequent work.

DLVR-M — Figure 4. DLVR-M particles efficiently deliver gene editors to a wide variety of transformed and primary cells

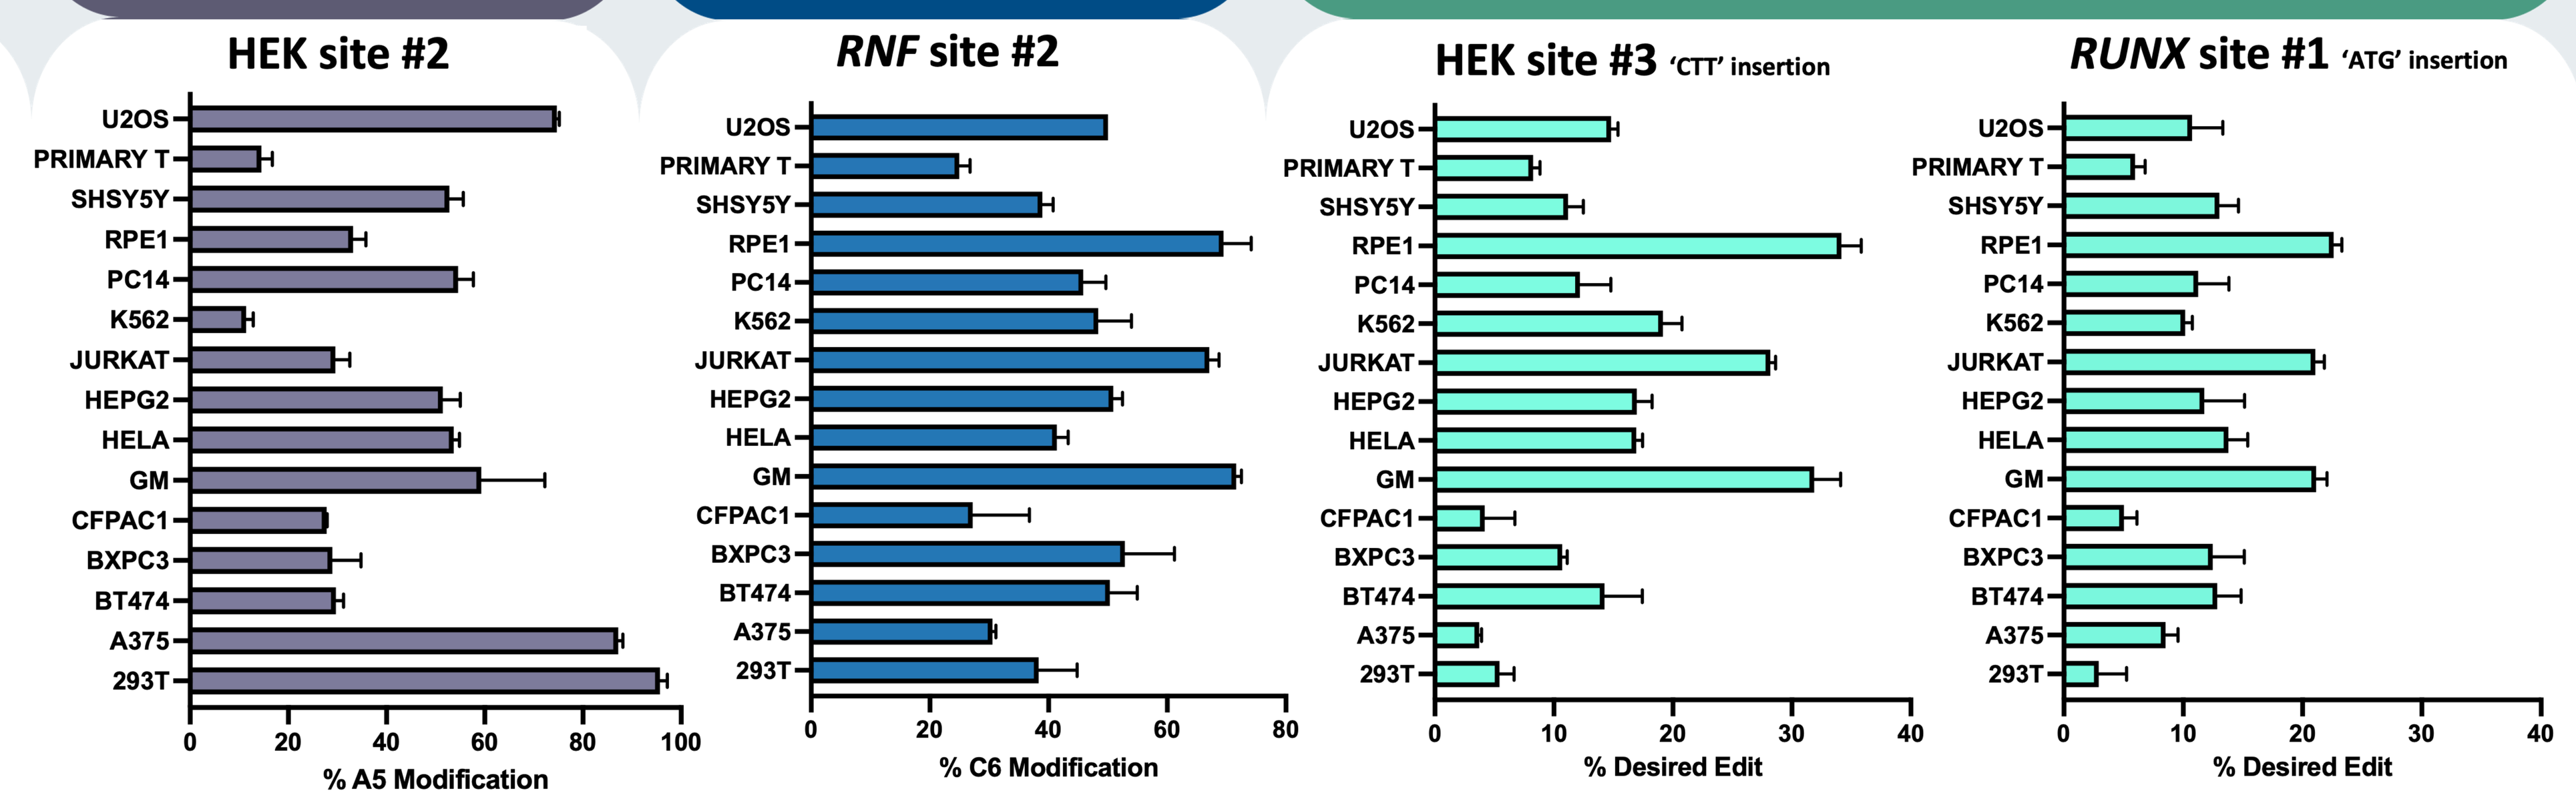

ABE MAX
BE4 MAX
Prime Editor (PE3)
HEK site #2
RNF site #2
HEK site #3 'CTT' insertion
RUNX site #1 'ATG' insertion
U2OS
PRIMARY T
SHSY5Y
RPE1
PC14
K562
JURKAT
HEPG2
HELA
GM
CFPAC1
BXPC3
BT474
A375
293T
% A5 Modification
% C6 Modification
% Desired Edit

DLVR-M (VSVG) particles can efficiently package and deliver Base[3,4] and Prime Editor[5] cargo into a variety of cells. DLVR-M particles do not require architecture modifications in order to efficiently deliver different types of gene editor RNP cargos. Particles were concentrated 100x by PEG precipitation prior to transduction of recipient cells.

DLVR-M *in vivo* — Figure 5. DLVR-M achieves high base editing efficiency in mouse liver with low editing in spleen

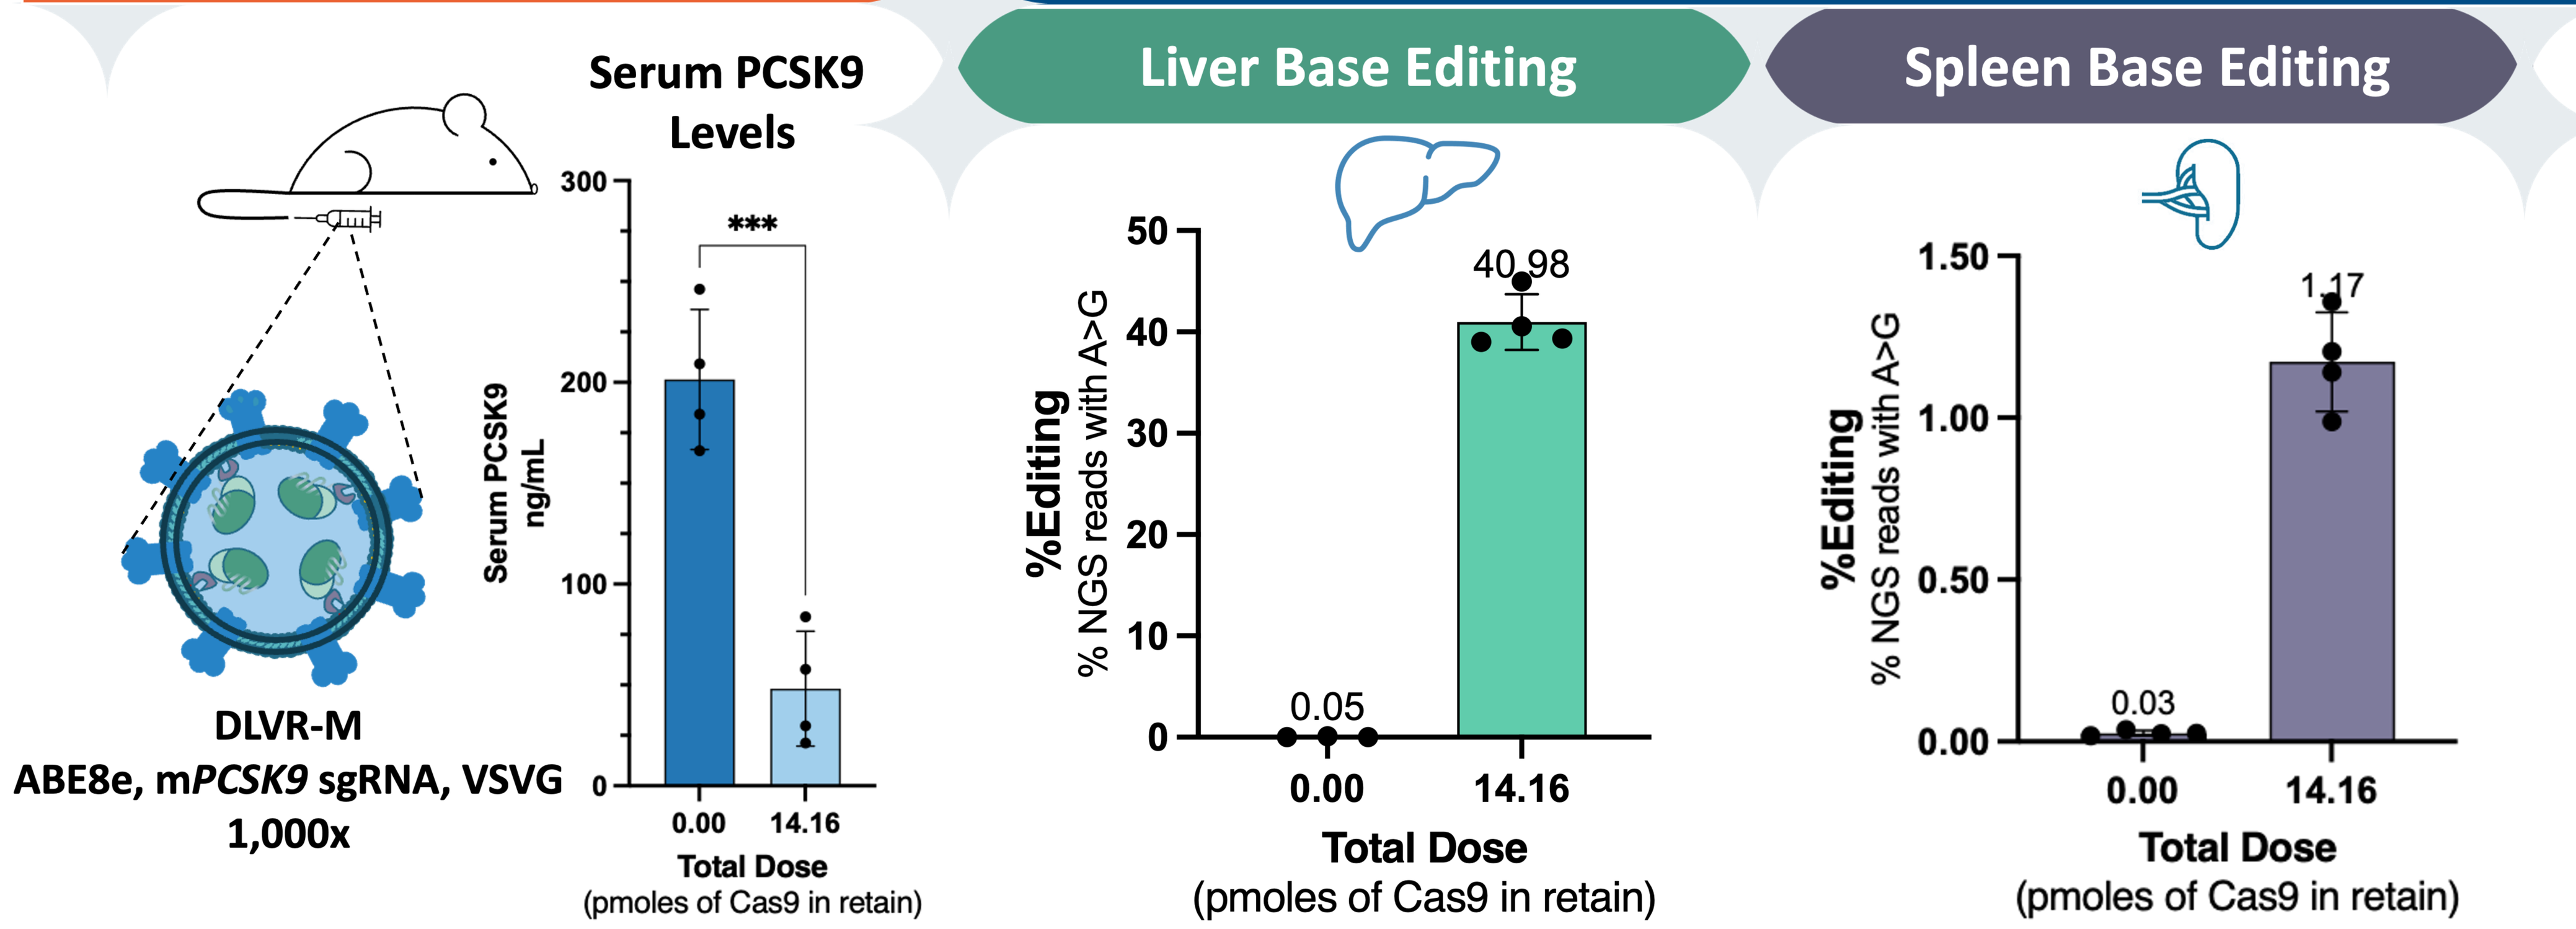

DLVR-M
ABE8e, mPCSK9 sgRNA, VSVG
1,000x
Serum PCSK9 Levels
Serum PCSK9 ng/mL

Liver Base Editing
Spleen Base Editing
40.98
0.05
1.17
0.03
%Editing % NGS reads with A>G
0.00
14.16
Total Dose (pmoles of Cas9 in retain)

The high efficiency bulk liver *PCSK9* editing only required 14 pmol of ABE8e as measured by Cas9 ELISA and yielded an approximate 75% reduction in serum PCSK9.

Humanized DLVR-M — Figure 6. Completely humanized DLVR-M efficiently delivers base editor RNP into primary human cells

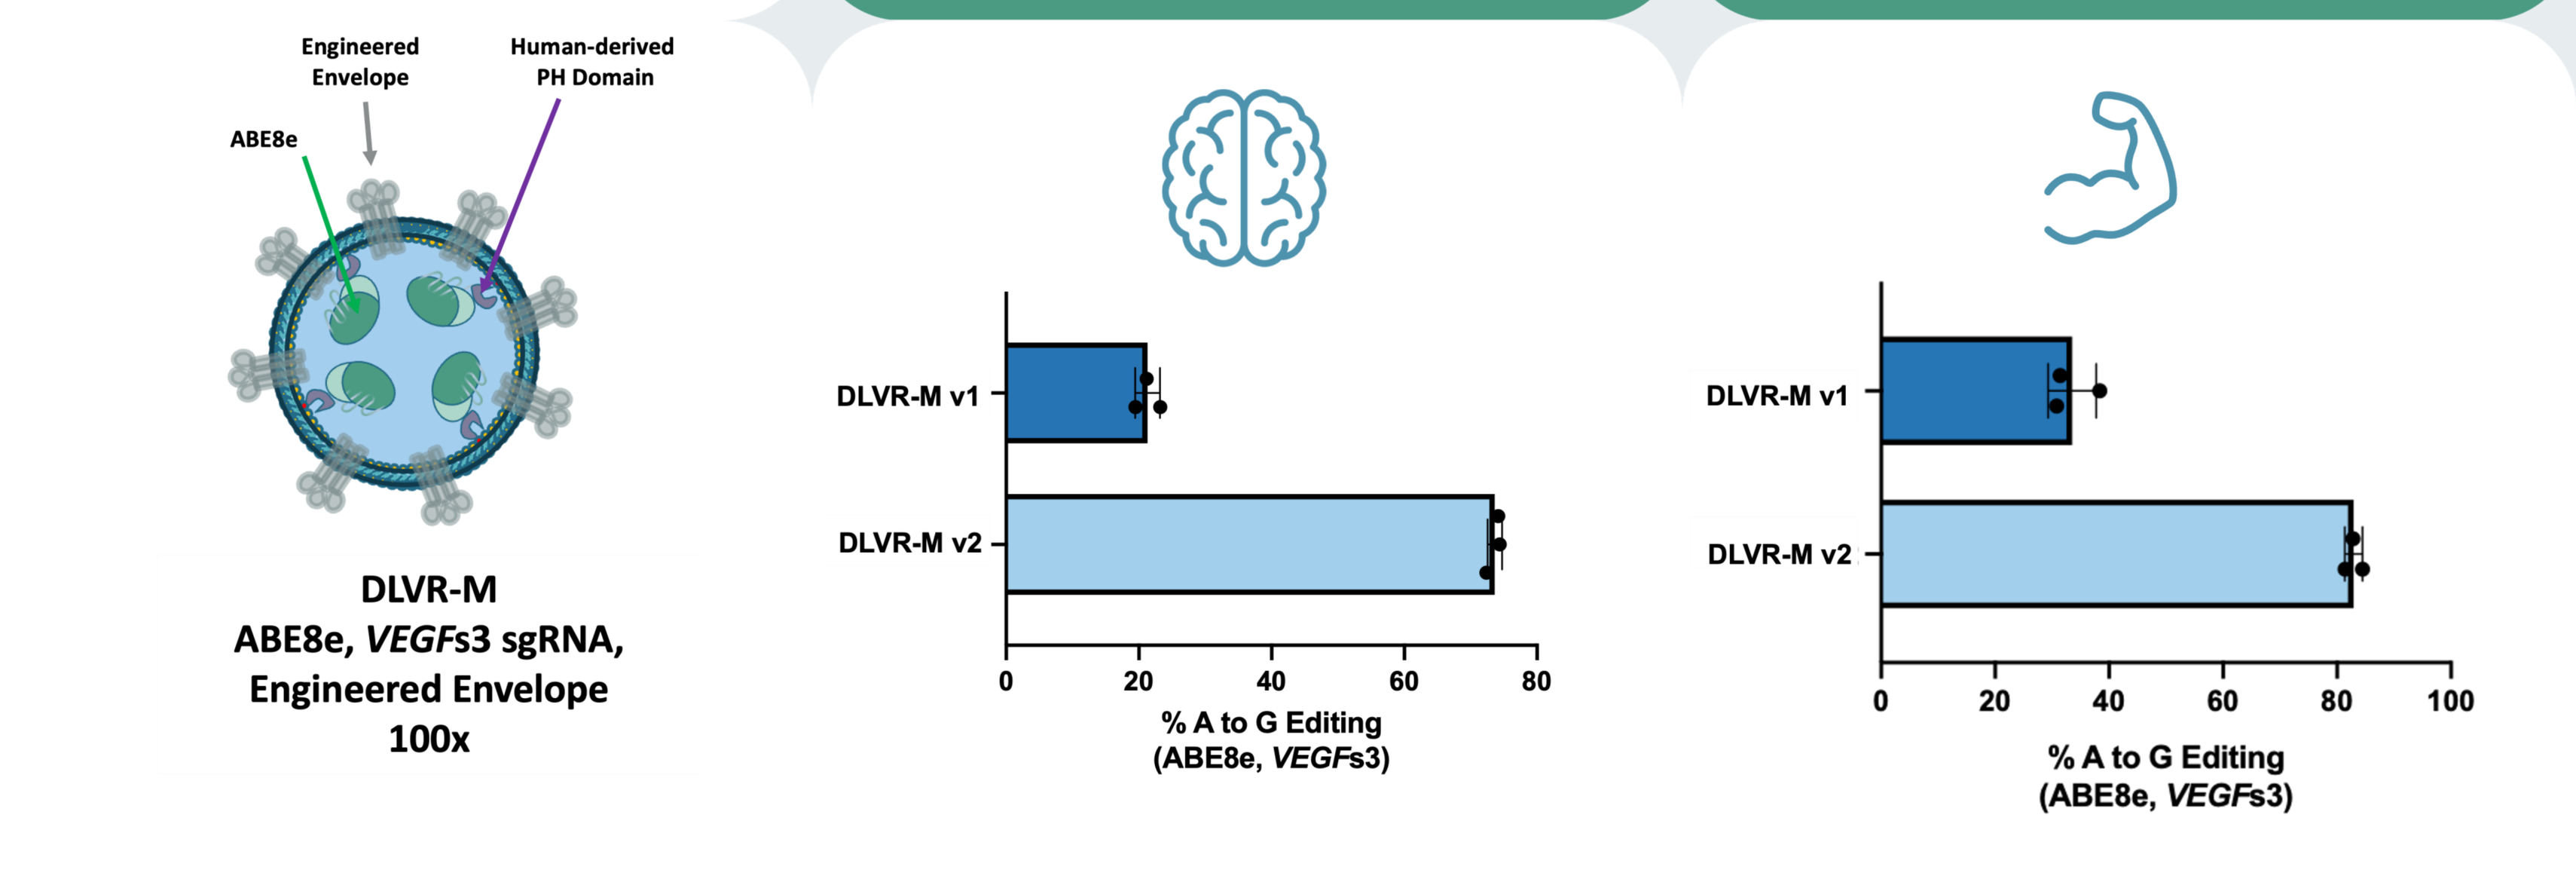

Primary Human Cortical Neurons
Primary Human Skeletal Muscle
Engineered Envelope
Human-derived PH Domain
ABE8e
DLVR-M
ABE8e, VEGFs3 sgRNA,
Engineered Envelope
100x
DLVR-M v1
DLVR-M v2
% A to G Editing (ABE8e, VEGFs3)

DLVR-M is capable of utilizing an envelope that has been engineered and humanized. Therefore, this DLVR-M particle is devoid of virus-derived components. An optimized PH domain is utilized for packaging and offloading of cargo. The DLVR-M v2 particle incorporates improvements in envelope presentation on the particle surface. Particles were concentrated 100x by PEG precipitation prior to transduction of recipient cells.

Humanized DLVR-M — Figure 7. Completely humanized DLVR-M efficiently delivers base editor RNP into unstimulated HSPCs

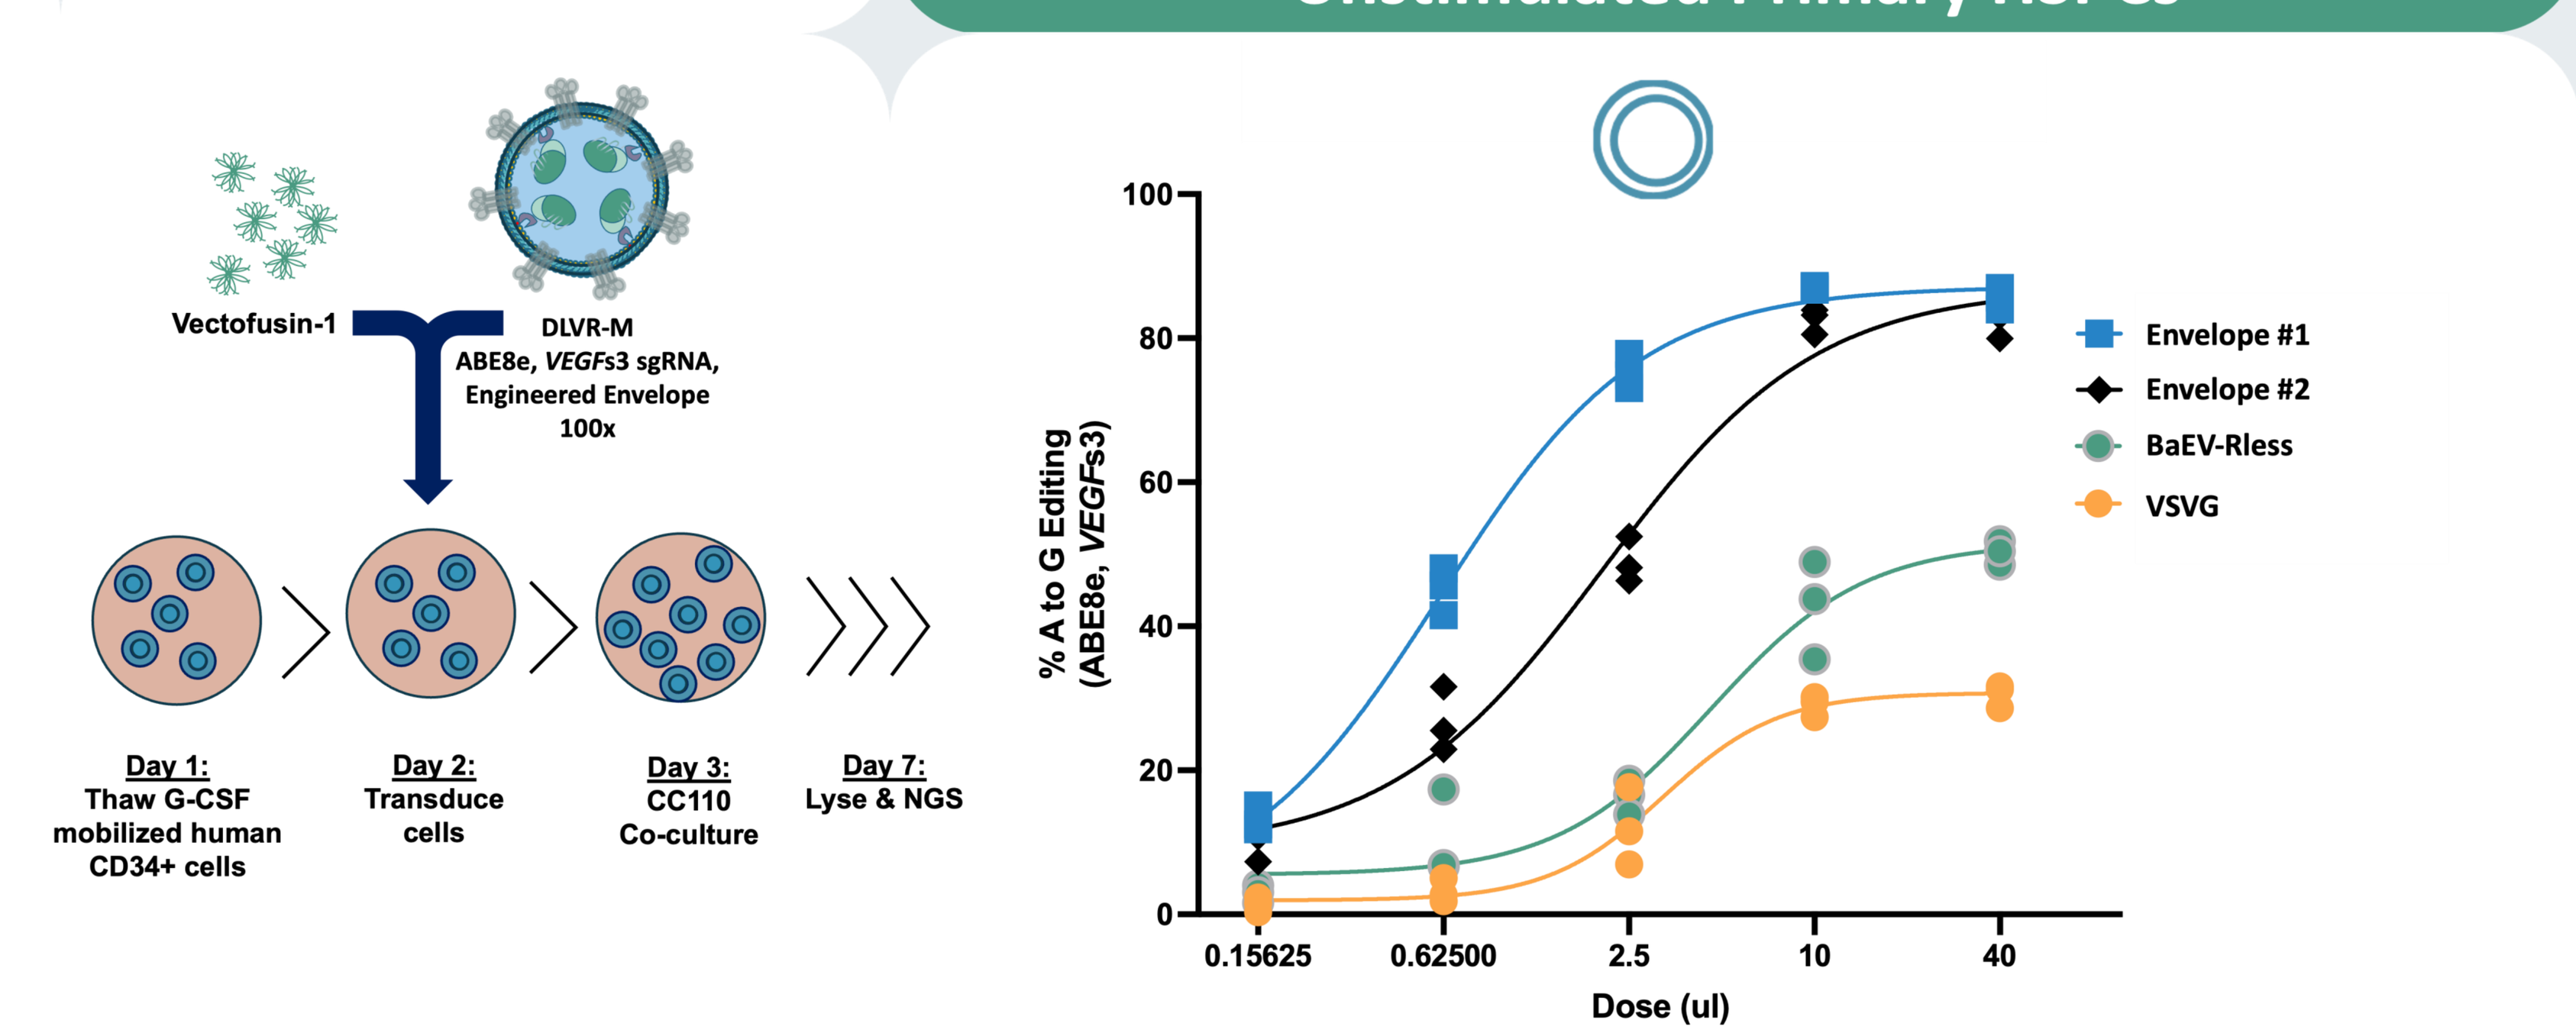

Unstimulated Primary HSPCs
Vectofusin-1
DLVR-M
ABE8e, VEGFs3 sgRNA,
Engineered Envelope
100x
Day 1: Thaw G-CSF mobilized human CD34+ cells
Day 2: Transduce cells
Day 3: CC110 Co-culture
Day 7: Lyse & NGS
% A to G Editing (ABE8e, VEGFs3)
Dose (ul)
Envelope #1
Envelope #2
BaEV-Rless
VSVG

DLVR-M novel, humanized envelopes outperform VSVG-pseudotyped DLVR-M particles. Particles were concentrated 100x by PEG precipitation and mixed with Vectofusin-1[6] prior to transduction of unstimulated primary human hematopoietic stem and progenitor cells (HSPCs), as outlined above. CC110 is a serum-free culture supplement for expansion of human hematopoietic cells.

Conclusion

- The DLVR-M platform provides new capabilities for delivering a wide range of diverse protein and/or RNA cargos to a variety of different cell types with potentially reduced immunogenicity because envelope proteins are derived from and expressed in human cells.
- We anticipate that these new particles will be broadly useful for many research and therapeutics applications currently limited by the capabilities and characteristics of existing delivery modalities.
- This work introduces novel envelopes and showcases the DLVR-M platform capabilities and potential for efficient delivery of macromolecules *in vivo* and in therapeutically relevant primary cells which are typically not receptive to conventional delivery modalities.
- Future work will include a more detailed characterization of physical properties and immunological features of these novel particles.

Acknowledgements

We would like to express our gratitude to the patients who donated T cells, and Drs. Marcela Maus, Stephanie Renae Bailey and Andrea Schmidts for kindly providing primary T cells for this study. We owe a special thanks to Dr. Mahmudul Hasan for helpful discussion and NGS support. Finally, we thank Dr. Luca Biasco for insights related to HSPC experiments.

References

1. Balla T & Várnai P. Sci STKE. 2002. doi: 10.1126/stke.2002.125.pl3. PMID: 11917154.
2. Mangeot PE, et al. Nat Commun. 2019. doi: 10.1038/s41467-018-07845-z. PMID: 30604748
3. Komor AC, et al. Sci Adv. 2017. doi: 10.1126/sciadv.aao4774. PMID: 28875174.
4. Gaudelli NM, et al. Nature. 2017. doi: 10.1038/nature24644. PMID: 29160308.
5. Anzalone AV, et al. Nature. 2019 . doi: 10.1038/s41586-019-1711-4. PMID: 31634902.
6. Fenard D, et al. Mol Ther Nucleic Acids. 2013. PMID: 23653154.

Confidential – Property of Nvelop Therapeutics